Development of a new workflow for multiple attribute methodology (MAM) of an antibody drug conjugate (ADC)

The Power of Precision

Armelle Martelet[1], Valérie Garrigue[1], Hélène Le Borgne[1], Zoe Zhang[2], Bruno Genet[1], Kerstin Pohl[2]

[1]Sanofi, France; [2]SCIEX, USA

INTRODUCTION

- There are a variety of post-translational modifications (PTMs) known to occur in monoclonal antibody and ADC biotherapeutics during the manufacturing, formulation, and storage process
- Monitoring these quality attributes is of major interest because of the potential impact on a product's safety and efficacy
- Peptide mapping analysis using liquid chromatography mass spectrometry (LC-MS) based detection is commonly used for the identification and the relative quantification of attributes such as e.g. oxidation and deamidation
- However, establishing a workflow for streamlined data analysis can be challenging for stability and especially forced degradation studies with increasing numbers of samples.

MATERIALS AND METHODS

For forced degradation assessment ADC samples underwent thermal, mechanical and chemical stresses. All samples, including controls, were reduced, alkylated and enzymatically digested. Obtained peptides were separated on a reversed-phase C18 column using a high-flow LC setup (ExionLC™ System). MS detection was carried out on a quadrupole-time-of-flight instrument (SCIEX X500B QTOF System) using data-dependent acquisition. Subsequent data analysis was performed using SCIEX OS Software 1.7 in a new streamlined way combining a high level of automation for calculations, ease of review, and verification of the results.

Row	IS	Group	Name	Chemical Formula	Adduct/Ch...	Precursor (Q1) Mass (Da)	XIC Width (ppm)	Retention Time Mode	Retention Time (min)	Experiment Index
15	☐	LC			[M+2H]2+	899.94326	12.00076	RT value	57.70	1 +TOF MS (250 - 1800)
16	☐	LC			[M+3H]3+	600.29793	11.99404	RT value	57.70	1 +TOF MS (250 - 1800)
17	☐	LC			[M+2H]2+	899.94326	12.00076	RT value	62.67	1 +TOF MS (250 - 1800)
18	☐	LC			[M+3H]3+	600.29793	11.99404	RT value	62.67	1 +TOF MS (250 - 1800)
19	☐	LC			[M+2H]2+	899.94326	12.00076	RT value	63.94	1 +TOF MS (250 - 1800)
20	☐	LC			[M+3H]3+	600.29793	11.99404	RT value	64.02	1 +TOF MS (250 - 1800)
21	☐	LC			[M+3H]3+	1207.24129	11.99429	RT value	43.00	1 +TOF MS (250 - 1800)
22	☐	LC			[M+4H]4+	905.68279	12.00199	RT value	43.00	1 +TOF MS (250 - 1800)
23	☐	LC			[M+5H]5+	724.74769	12.00418	RT value	43.04	1 +TOF MS (250 - 1800)
24	☐	LC			[M+6H]6+	604.12429	12.00084	RT value	43.10	1 +TOF MS (250 - 1800)
25	☐	LC			[M+3H]3+	1207.24129	11.99429	RT value	42.02	1 +TOF MS (250 - 1800)
26	☐	LC			[M+4H]4+	905.68279	12.00199	RT value	42.02	1 +TOF MS (250 - 1800)
27	☐	LC			[M+3H]3+	1207.24129	11.99429	RT value	43.29	1 +TOF MS (250 - 1800)
28	☐	LC			[M+4H]4+	905.68279	12.00199	RT value	43.29	1 +TOF MS (250 - 1800)
29	☐	HC			[M+3H]3+	1344.98719	12.00011	RT value	67.27	1 +TOF MS (250 - 1800)
30	☐	HC			[M+4H]4+	1008.99221	12.00207	RT value	67.25	1 +TOF MS (250 - 1800)
31	☐	HC			[M+5H]5+	807.39522	12.00156	RT value	67.25	1 +TOF MS (250 - 1800)
32	☐	HC			[M+6H]6+	672.99723	5.36406	RT value	67.25	1 +TOF MS (250 - 1800)
33	☐	HC			[M+3H]3+	1350.31883	11.99717	RT value	63.57	1 +TOF MS (250 - 1800)

Figure 1. Summary of quality attributes used for tracking purposes. Oxidation, deamidation and isoaspartate formation were defined to be monitored as part of the degradation assessment. Up to four charge states from each peptides were considered in quantification.

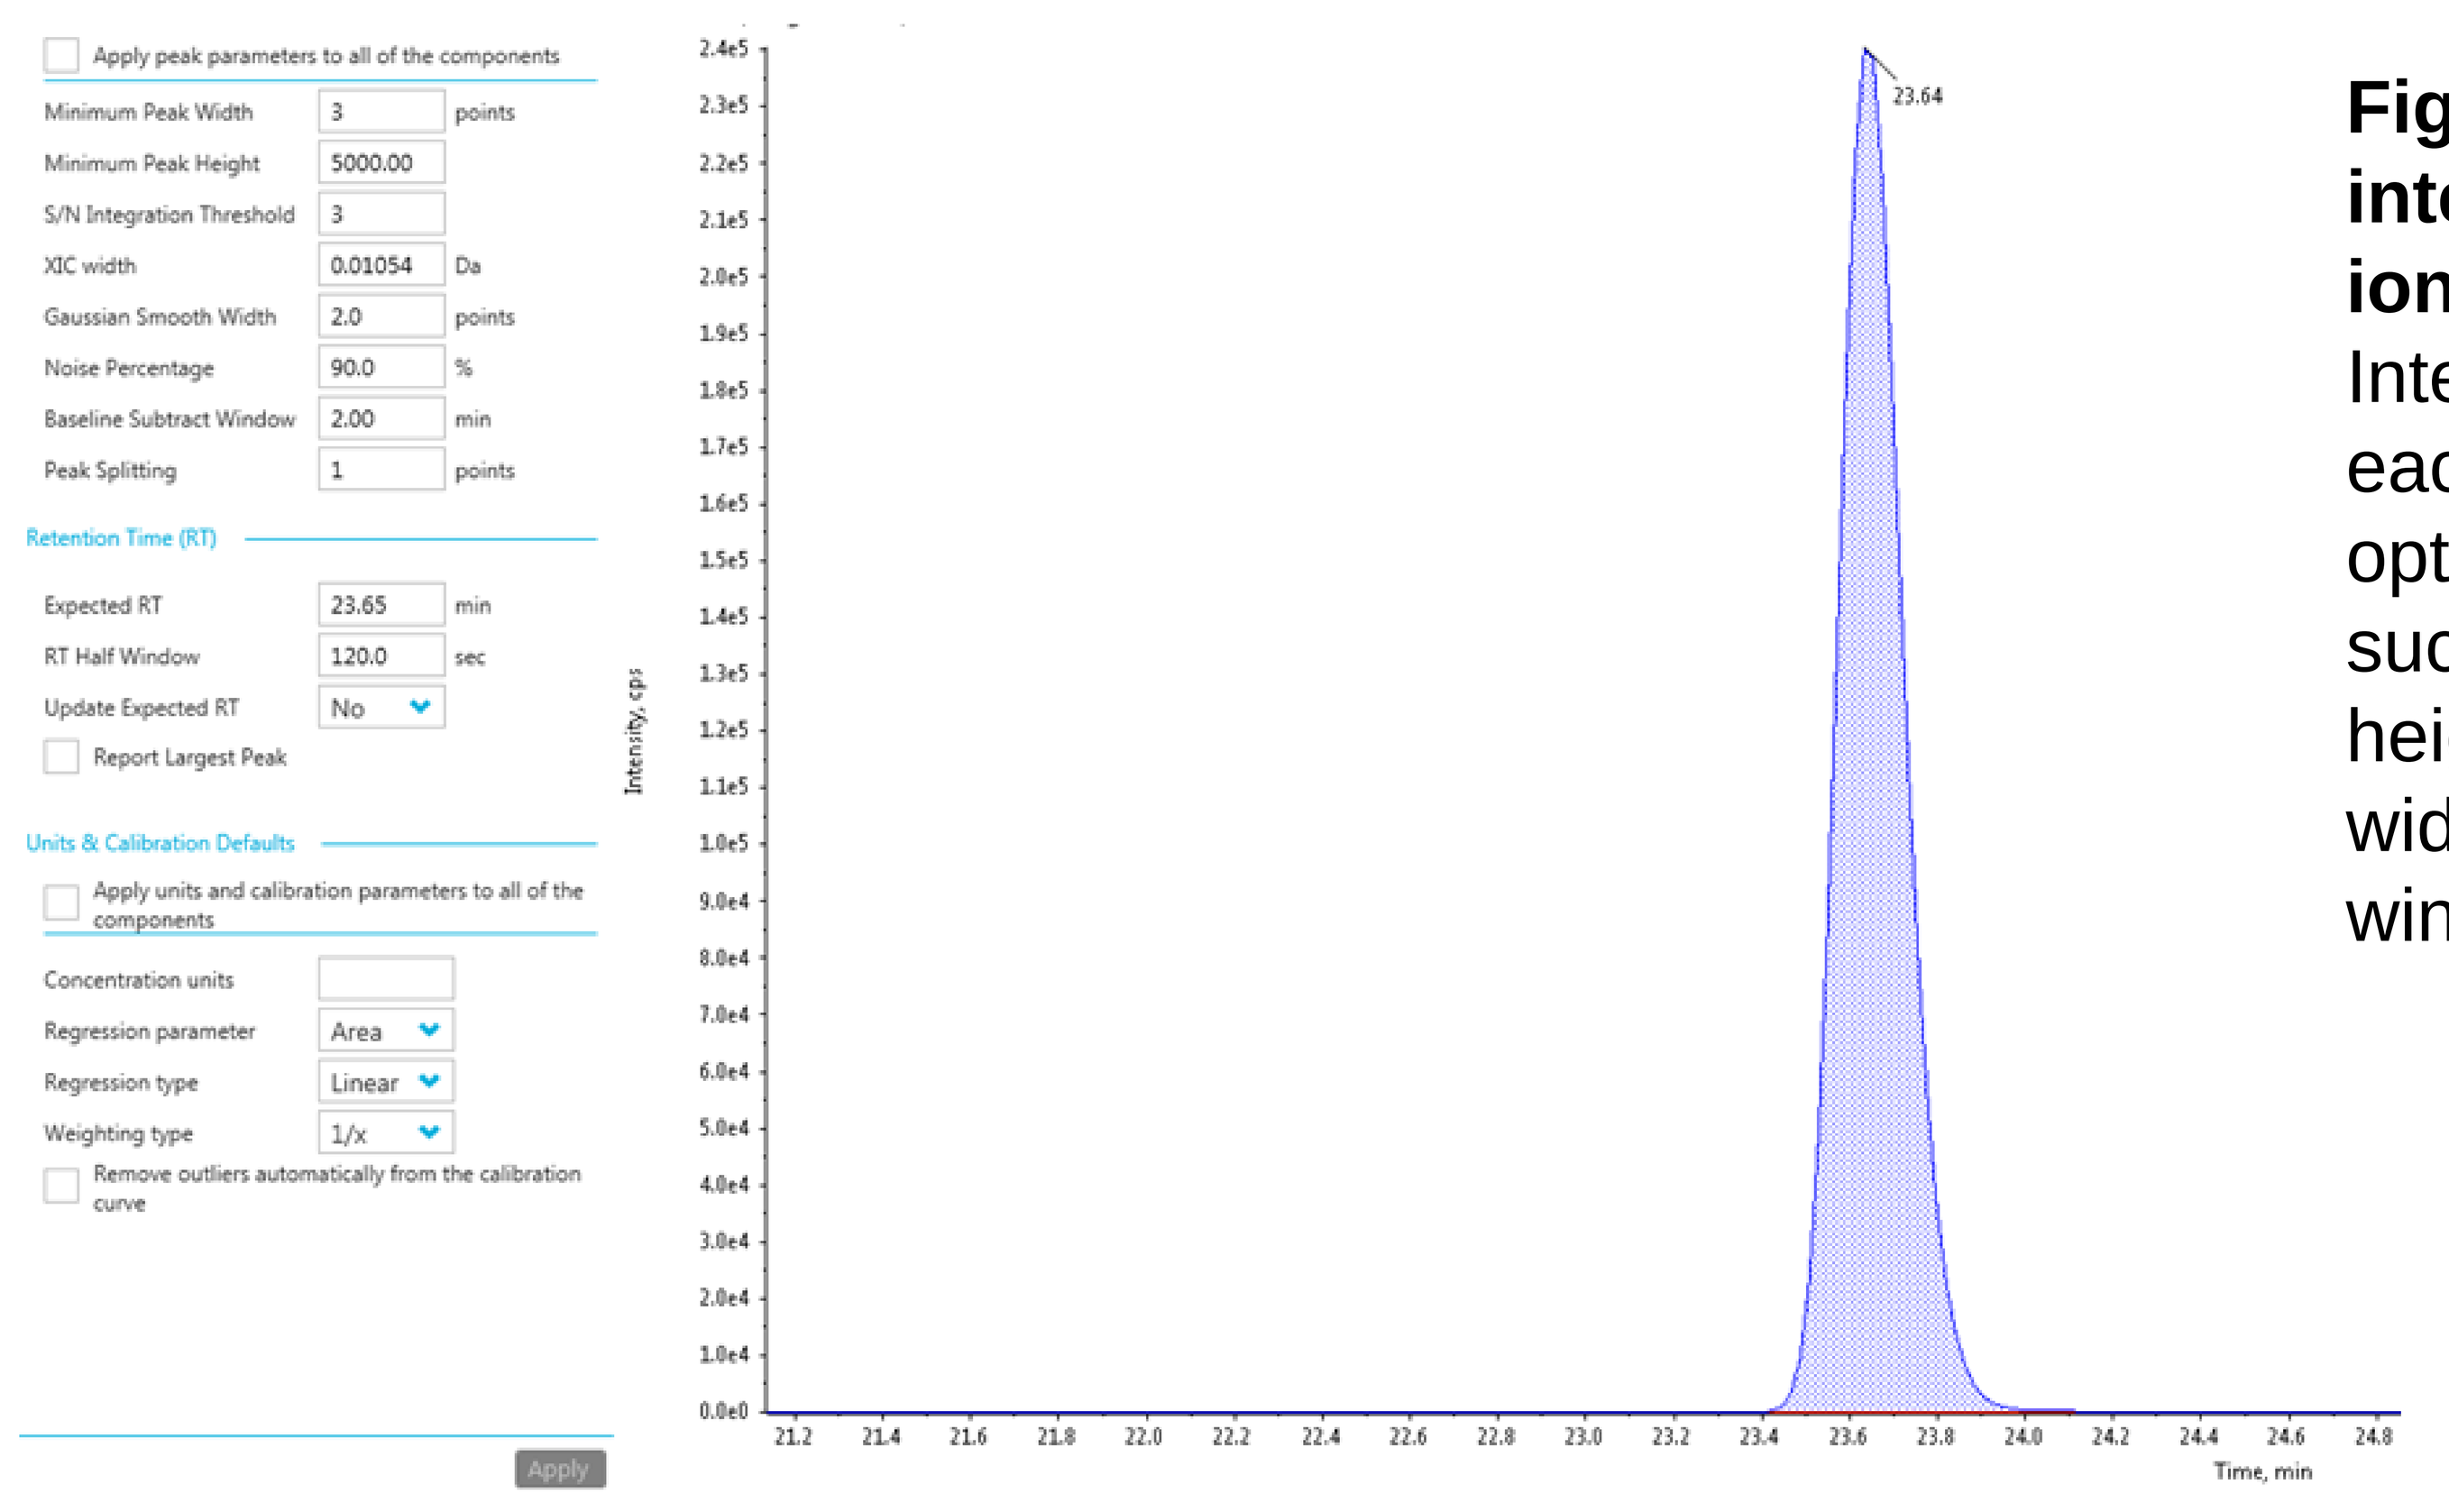

Figure 2. Example of the integration of an extracted ion chromatogram (XIC). Integration parameters for each attribute were optimized independently such as minimum peak height, S/N threshold, XIC width, peak splitting and RT window.

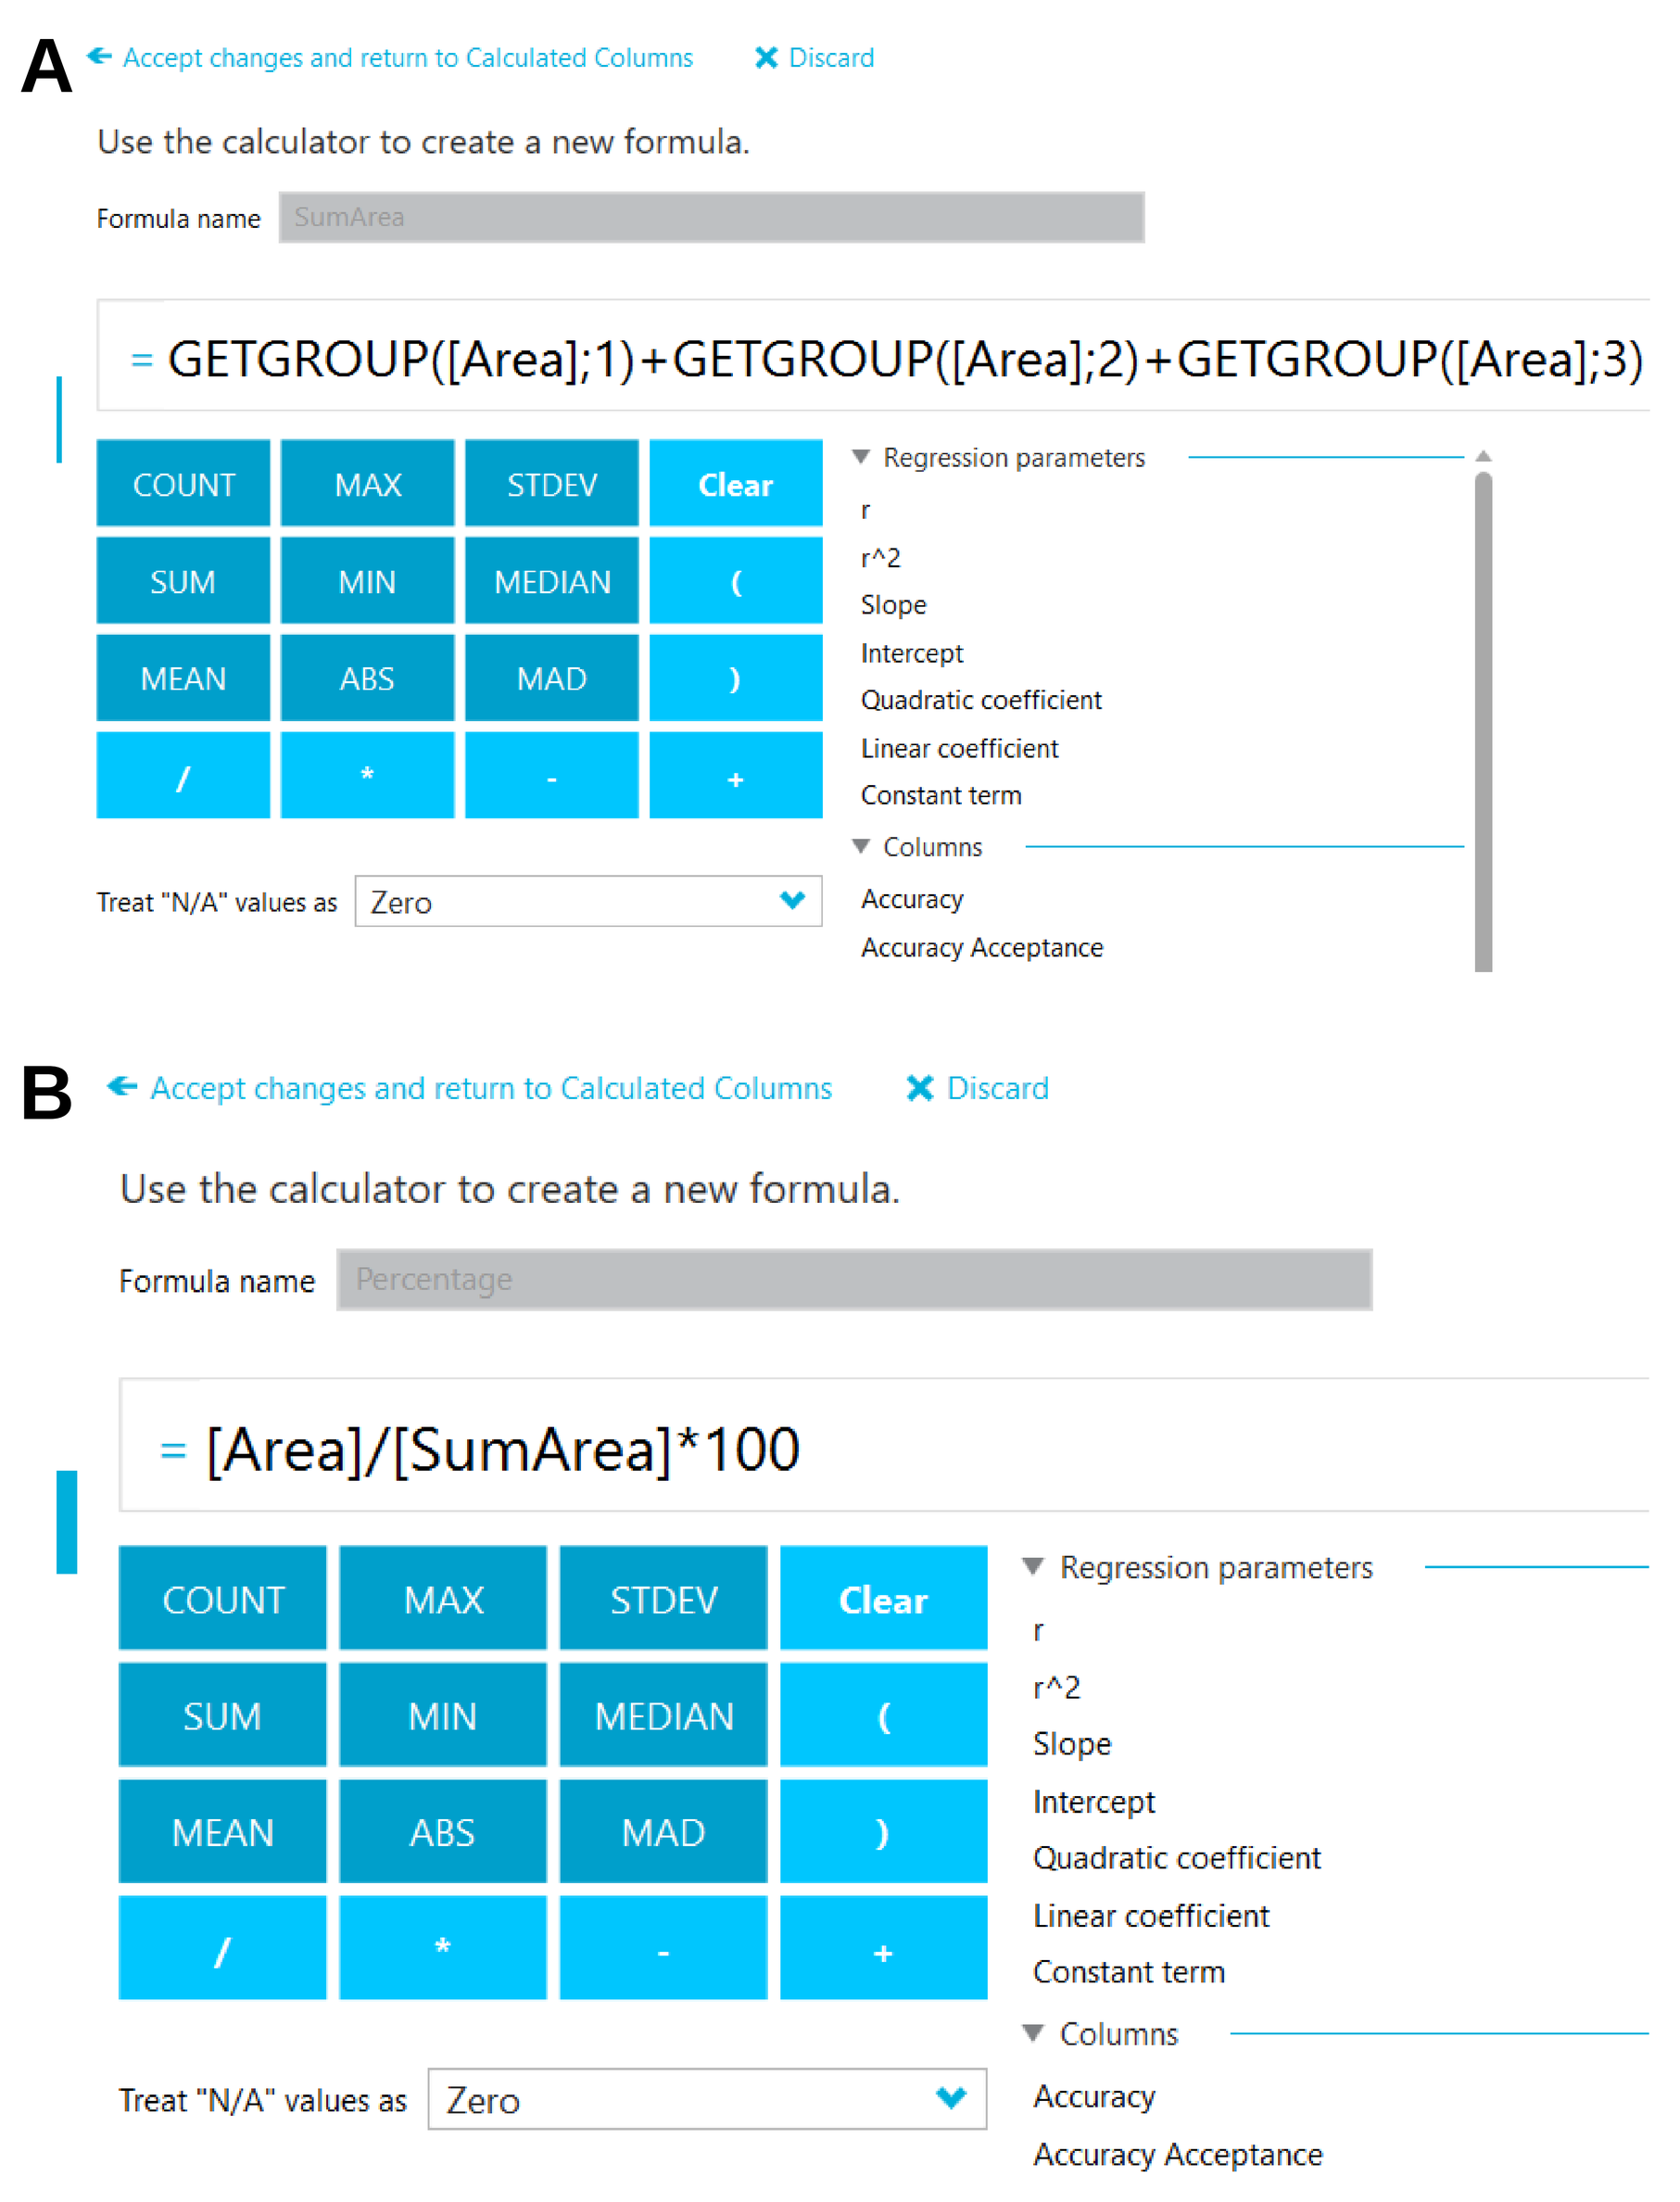
A
Use the calculator to create a new formula.
= GETGROUP([Area];1)+GETGROUP([Area];2)+GETGROUP([Area];3)
B
Use the calculator to create a new formula.
= [Area]/[SumArea]*100

Figure 3. User-defined calculations for automatic % calculation for each modification. An automatic calculation of modification percentages based on extracted ion chromatograms (XIC) areas of modified peptides against all other forms of the same peptide was set up. Up to four charge states were summed to achieve a precise calculation (A), followed by a percentage calculation (B).

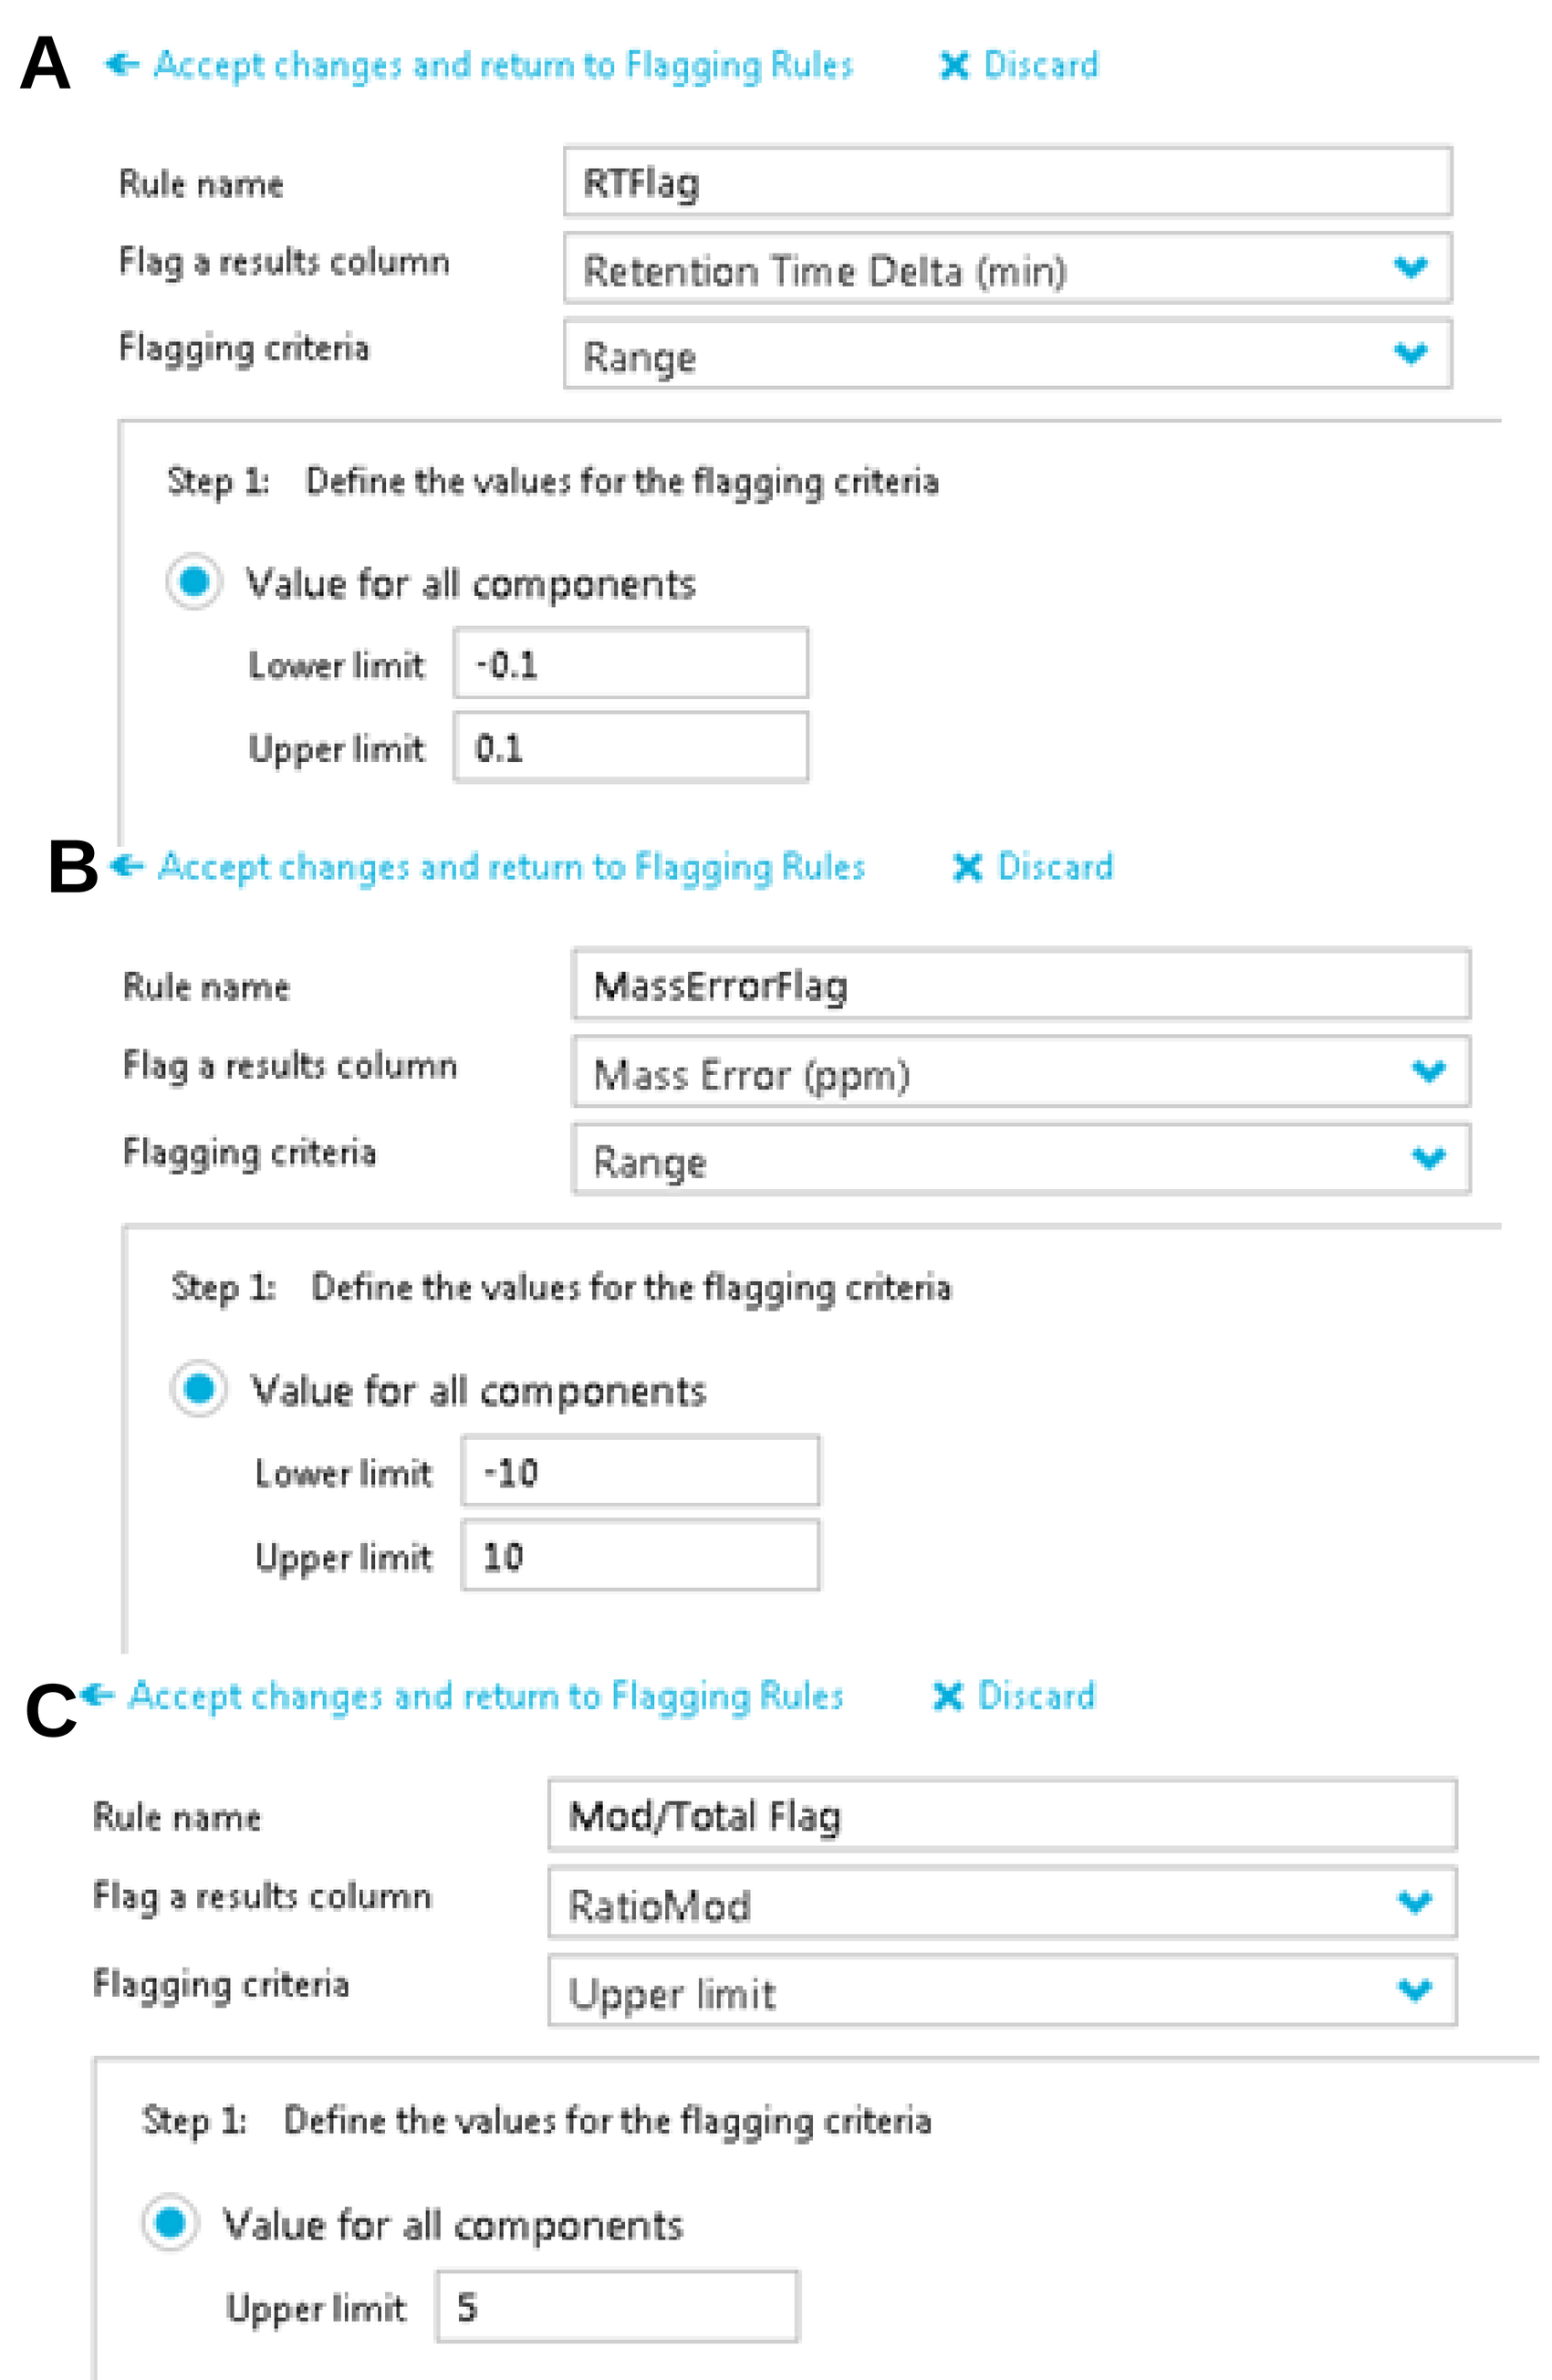
A
Rule name RTFlag
Flag a results column Retention Time Delta (min)
Flagging criteria Range
Lower limit -0.1
Upper limit 0.1
B
Rule name MassErrorFlag
Flag a results column Mass Error (ppm)
Flagging criteria Range
Lower limit -10
Upper limit 10
C
Rule name Mod/Total Flag
Flag a results column RatioMod
Flagging criteria Upper limit
Upper limit 5

Figure 4. Flagging rules for different parameters within MAM assay. To achieve an accurate identification and high throughput, attribute pass/fail criteria were defined for retention time (A), mass accuracy (B) and modification percentage (C). Any attribute which fails the set criteria will be flagged in the assay table by a color scheme, facilitating data review.

RESULTS

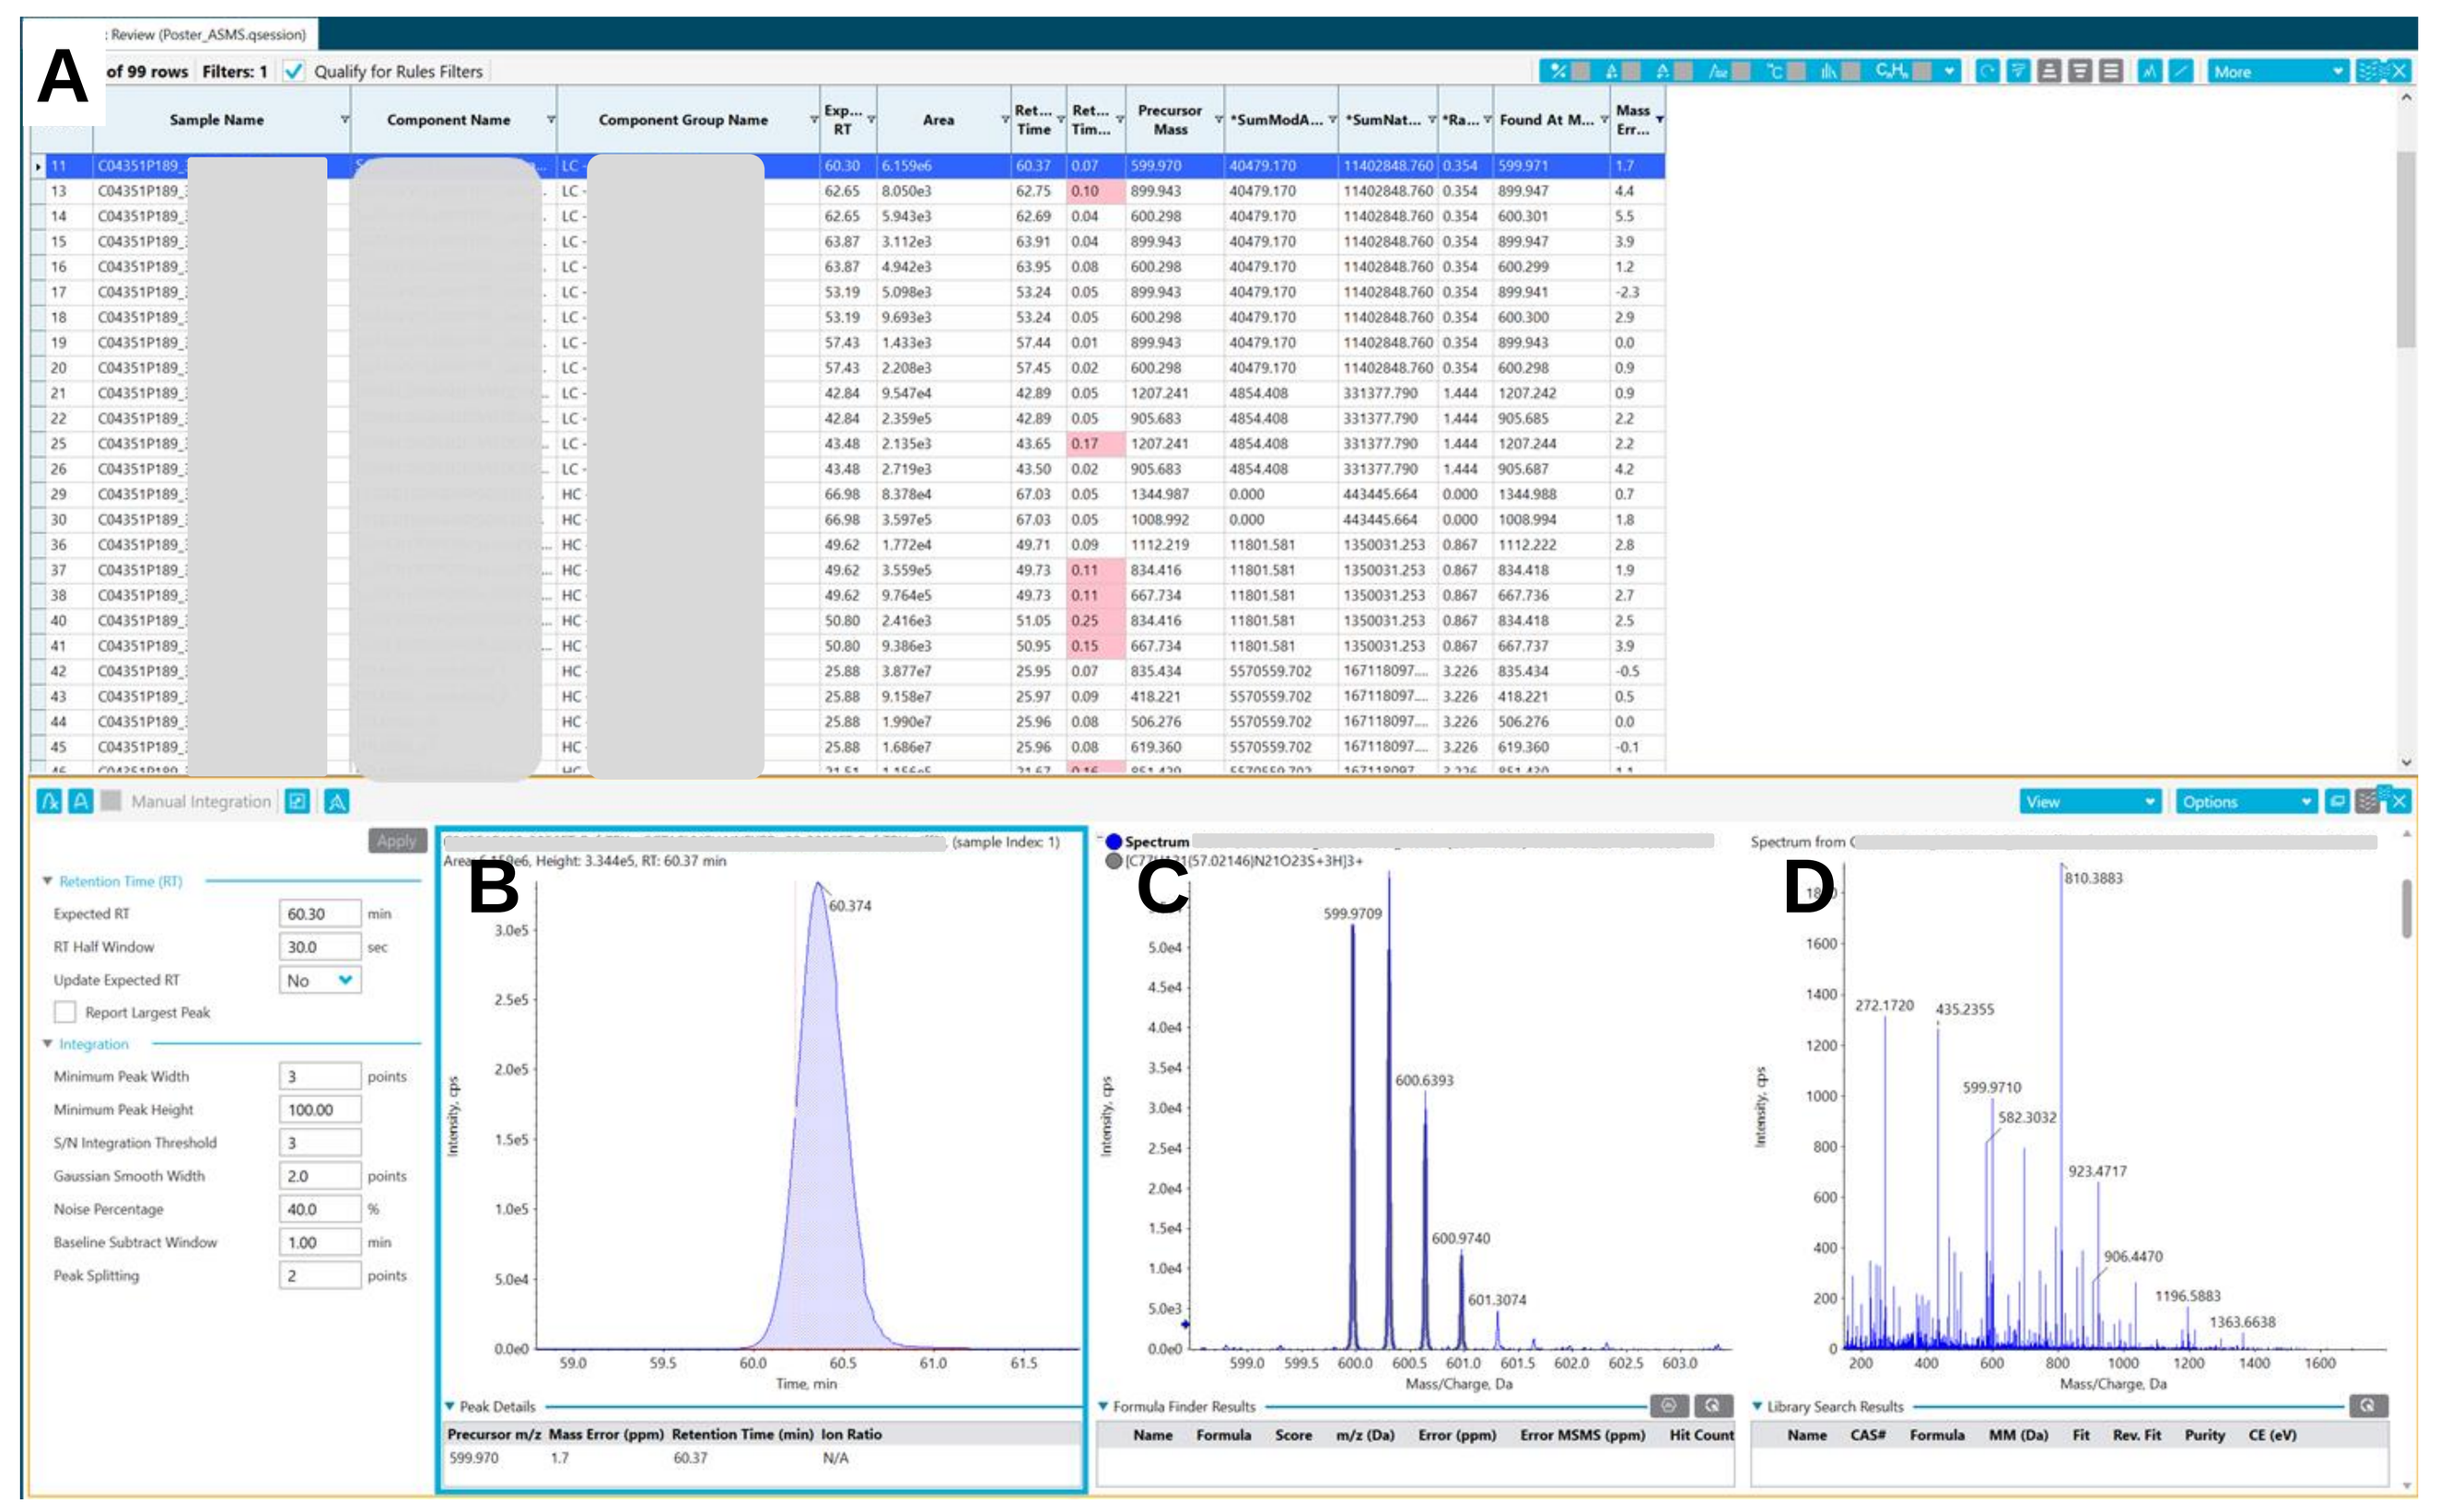

Figure 5. Results of the final MAM assay for the ADC stress study. Summary of all attributes in a table format (A). For each attribute XIC (B), underlying MS (C) and MS/MS (D) information is available. An overlay of the MS information with the theoretical isotope patterns provides full confidence in the assignment (gray pattern in C). Attributes failing the set criteria (red highlights) were interrogated further.

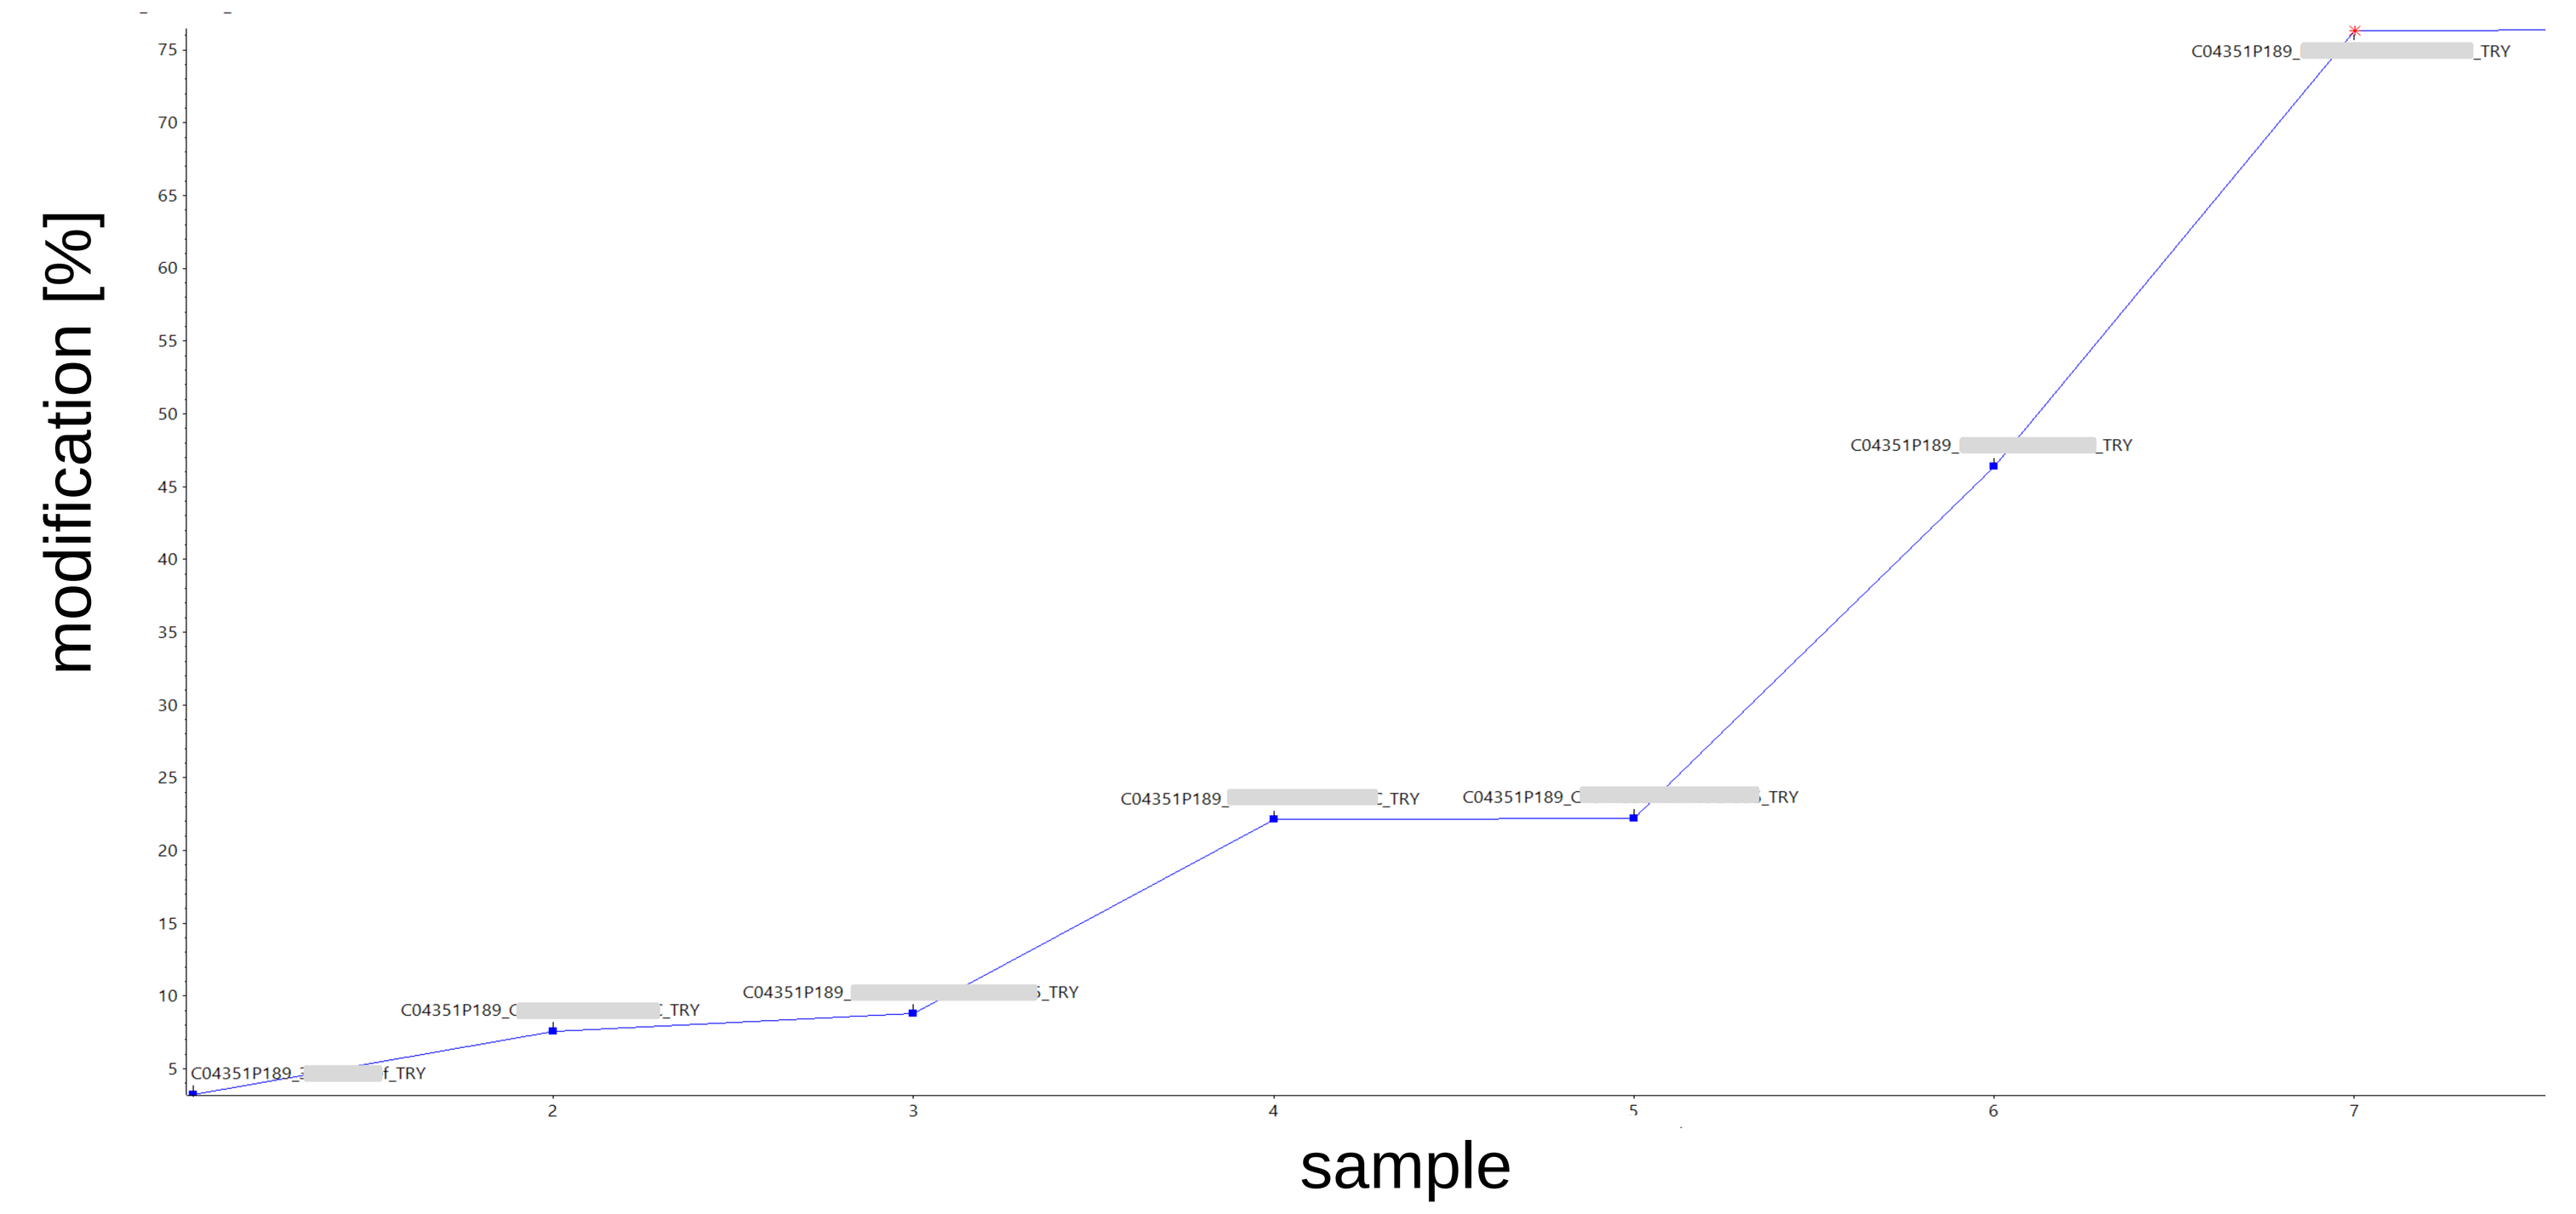
modification [%]
sample

Figure 6. Oxidation change on the peptide DTLMISR. Metric plot of the modification percentage change across stressed samples showing increasing levels of oxidation with increasing stress time. The metric plot can be directly generated inside the results table as a visualization tool for easy and quick comparison of samples.

CONCLUSIONS

- The established MAM assay represents a comprehensive solution for monitoring product quality attributes (PQA)
- A very flexible and automatic calculation of the percentage of modification including visual tools (metric plot, flagging of out-of-range parameters) speeds up the assessment of PQAs
- The streamlined workflow is compliant-ready, thus can be transferred to departments with further regulatory requirements

TRADEMARKS/LICENSING

The SCIEX clinical diagnostic portfolio is For In Vitro Diagnostic Use. Rx Only. Product(s) not available in all countries. For information on availability, please contact your local sales representative or refer to https://sciex.com/diagnostics. All other products are For Research Use Only. Not for use in Diagnostic Procedures.
Trademarks and/or registered trademarks mentioned herein, including associated logos, are the property of AB Sciex Pte. Ltd. or their respective owners in the United States and/or certain other countries.
© 2020 DH Tech. Dev. Pte. Ltd. RUO-MKT-10-11573-B. AB SCIEX™ is being used under license.